EXPLORATORY OCULAR SURFACE DISTRIBUTION STUDIES OF AZITHROMYCIN FORMULATIONS BASED ON SEMIFLUORINATED ALKANES

Fischer K.[1], Grillenberger R.[1], Amar T.[2], Krösser S.[1]
[1] Novaliq GmbH, 69120 Heidelberg, Germany; [2] IRIS Pharma, 06610 La Gaude, France

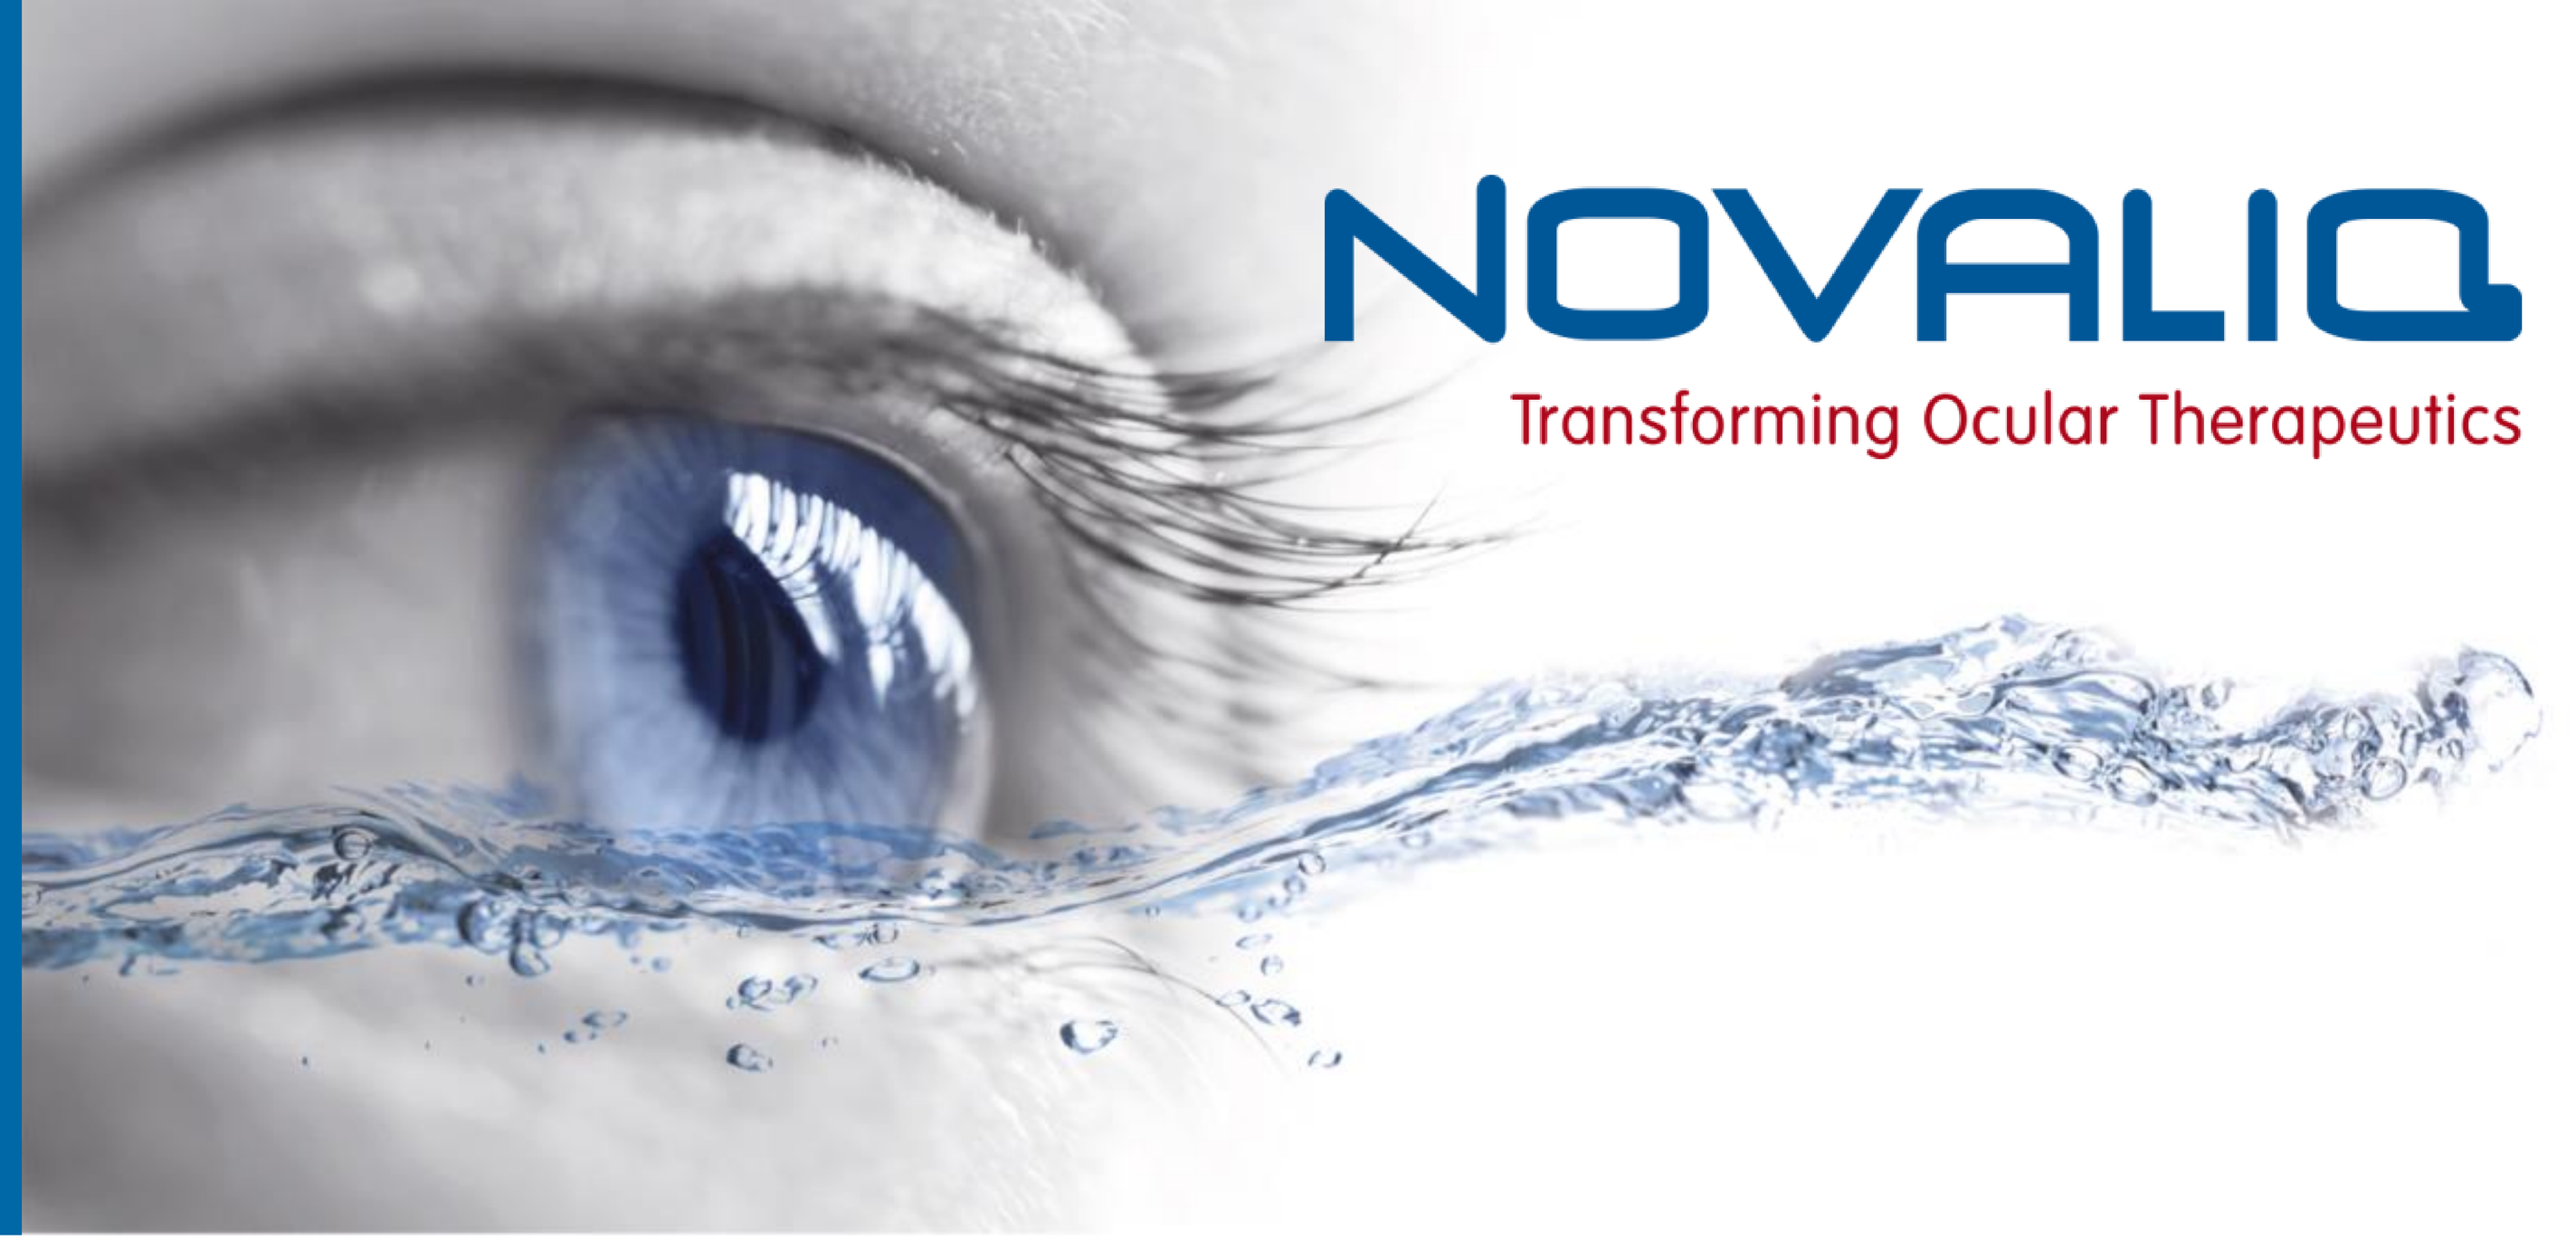
NOVALIQ
Transforming Ocular Therapeutics

Introduction and Purpose

Azithromycin is an antimicrobial agent with broad spectrum activity and anti-inflammatory properties. The aim of this study was to investigate the ocular distribution of azithromycin after a single topical instillation of a new semifluorinated alkane (SFA) based formulation in rabbits.

Methods

Rabbits were treated with a single administration of either 1.5% or 3% SFA-azithromycin suspensions. Due to the low surface and interface tension of the SFA, the droplet size is smaller compared to other ophthalmic formulations which lead to a low instillation volume of ~11 µL. Moreover, the spreading abilities of the drop are excellent. Tears, aqueous humor, cornea, bulbar conjunctiva, and eyelids from individual eyes were collected up to 144 hours post dosing and drug concentrations were measured using a validated RRLC-MS/MS method.

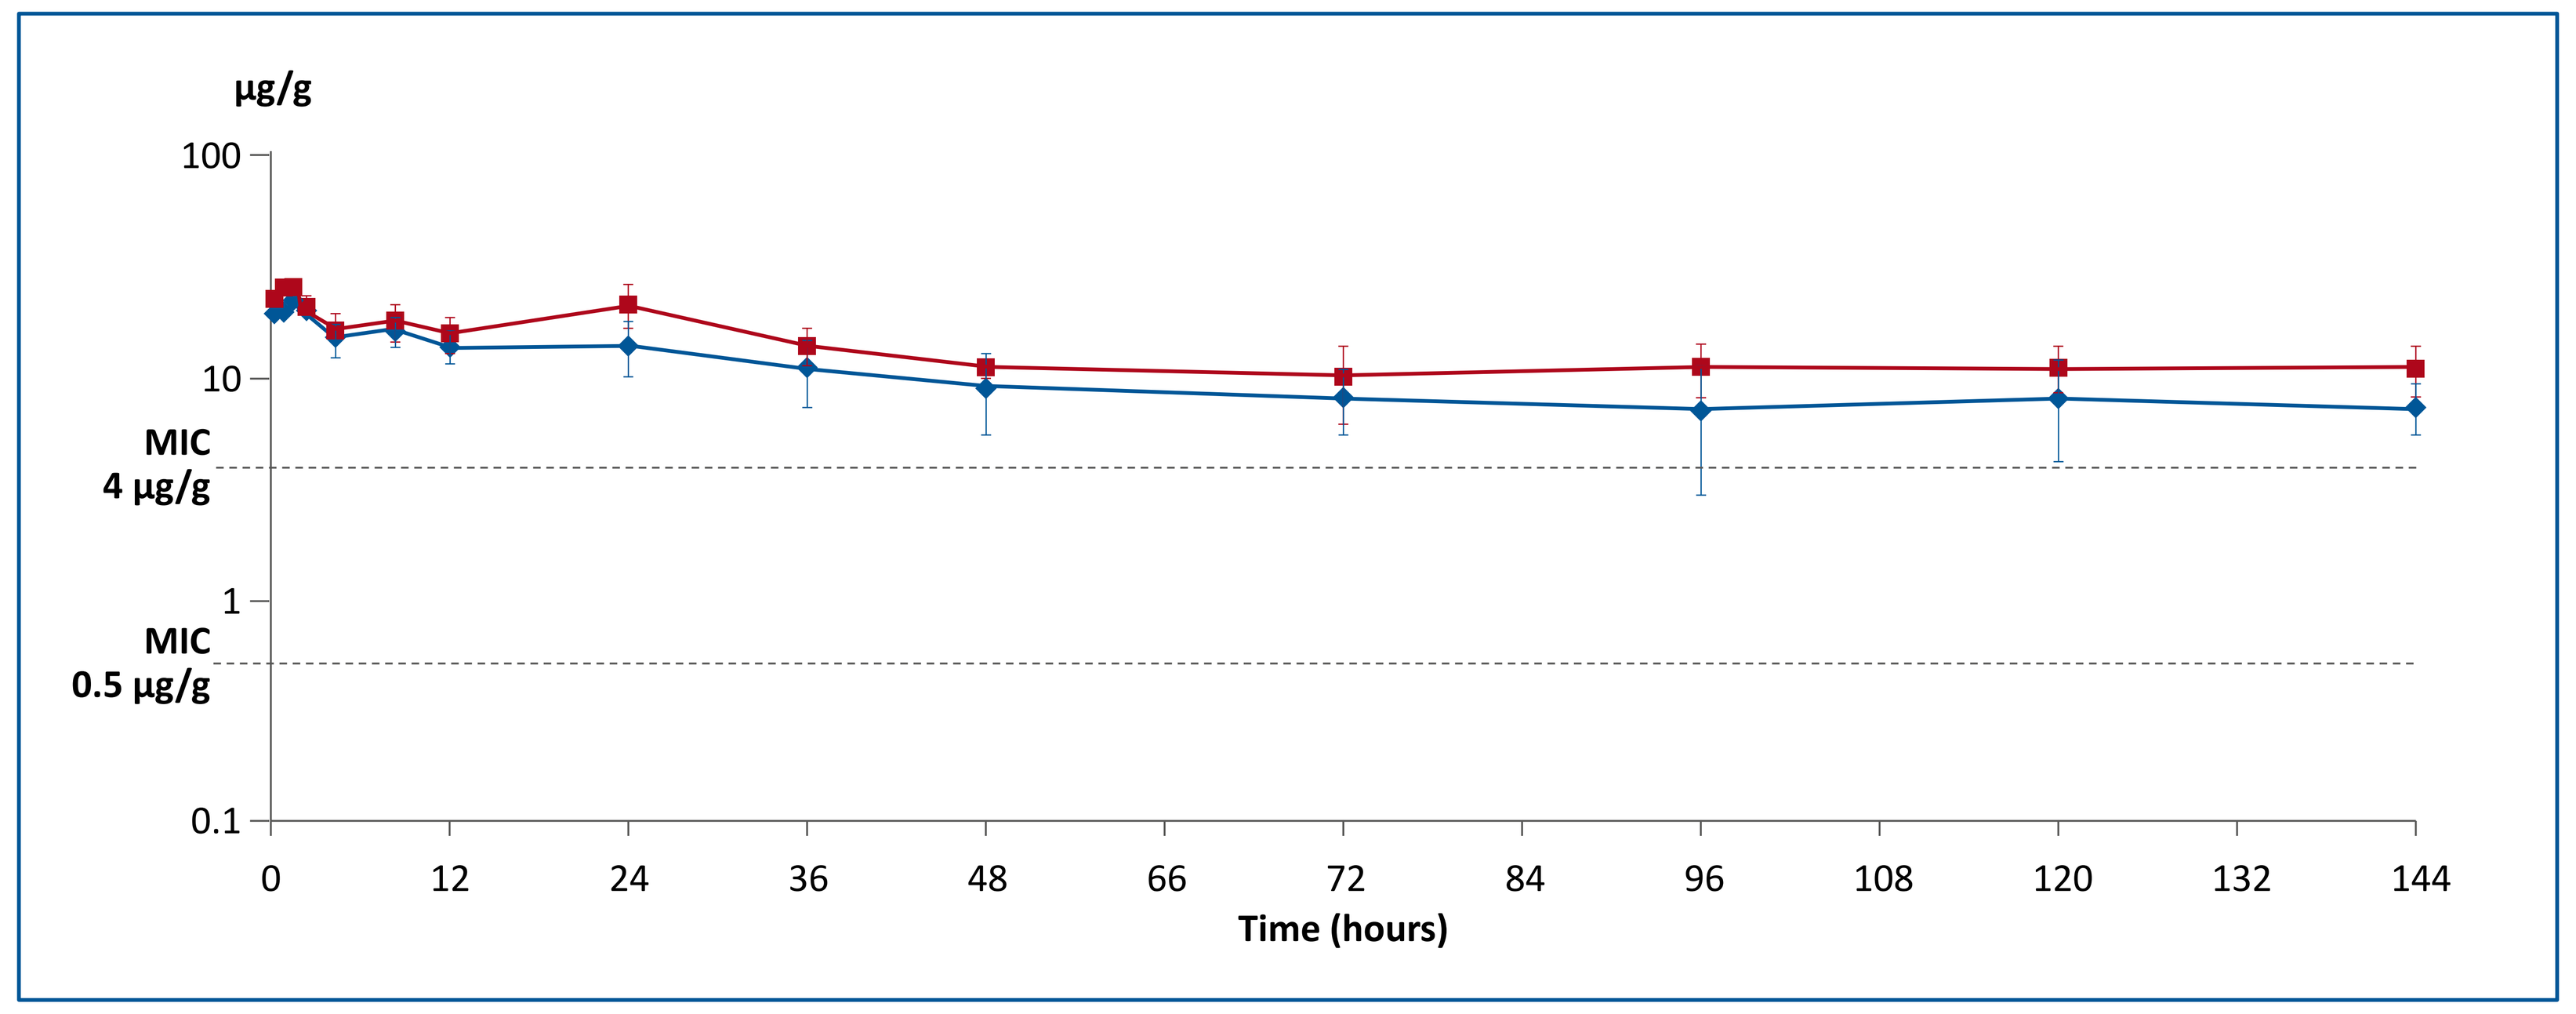
µg/g
100
10
MIC
4 µg/g
1
MIC
0.5 µg/g
0.1
0
12
24
36
48
66
72
84
96
108
120
132
144
Time (hours)

Figure 1: Azithromycin content in eyelid

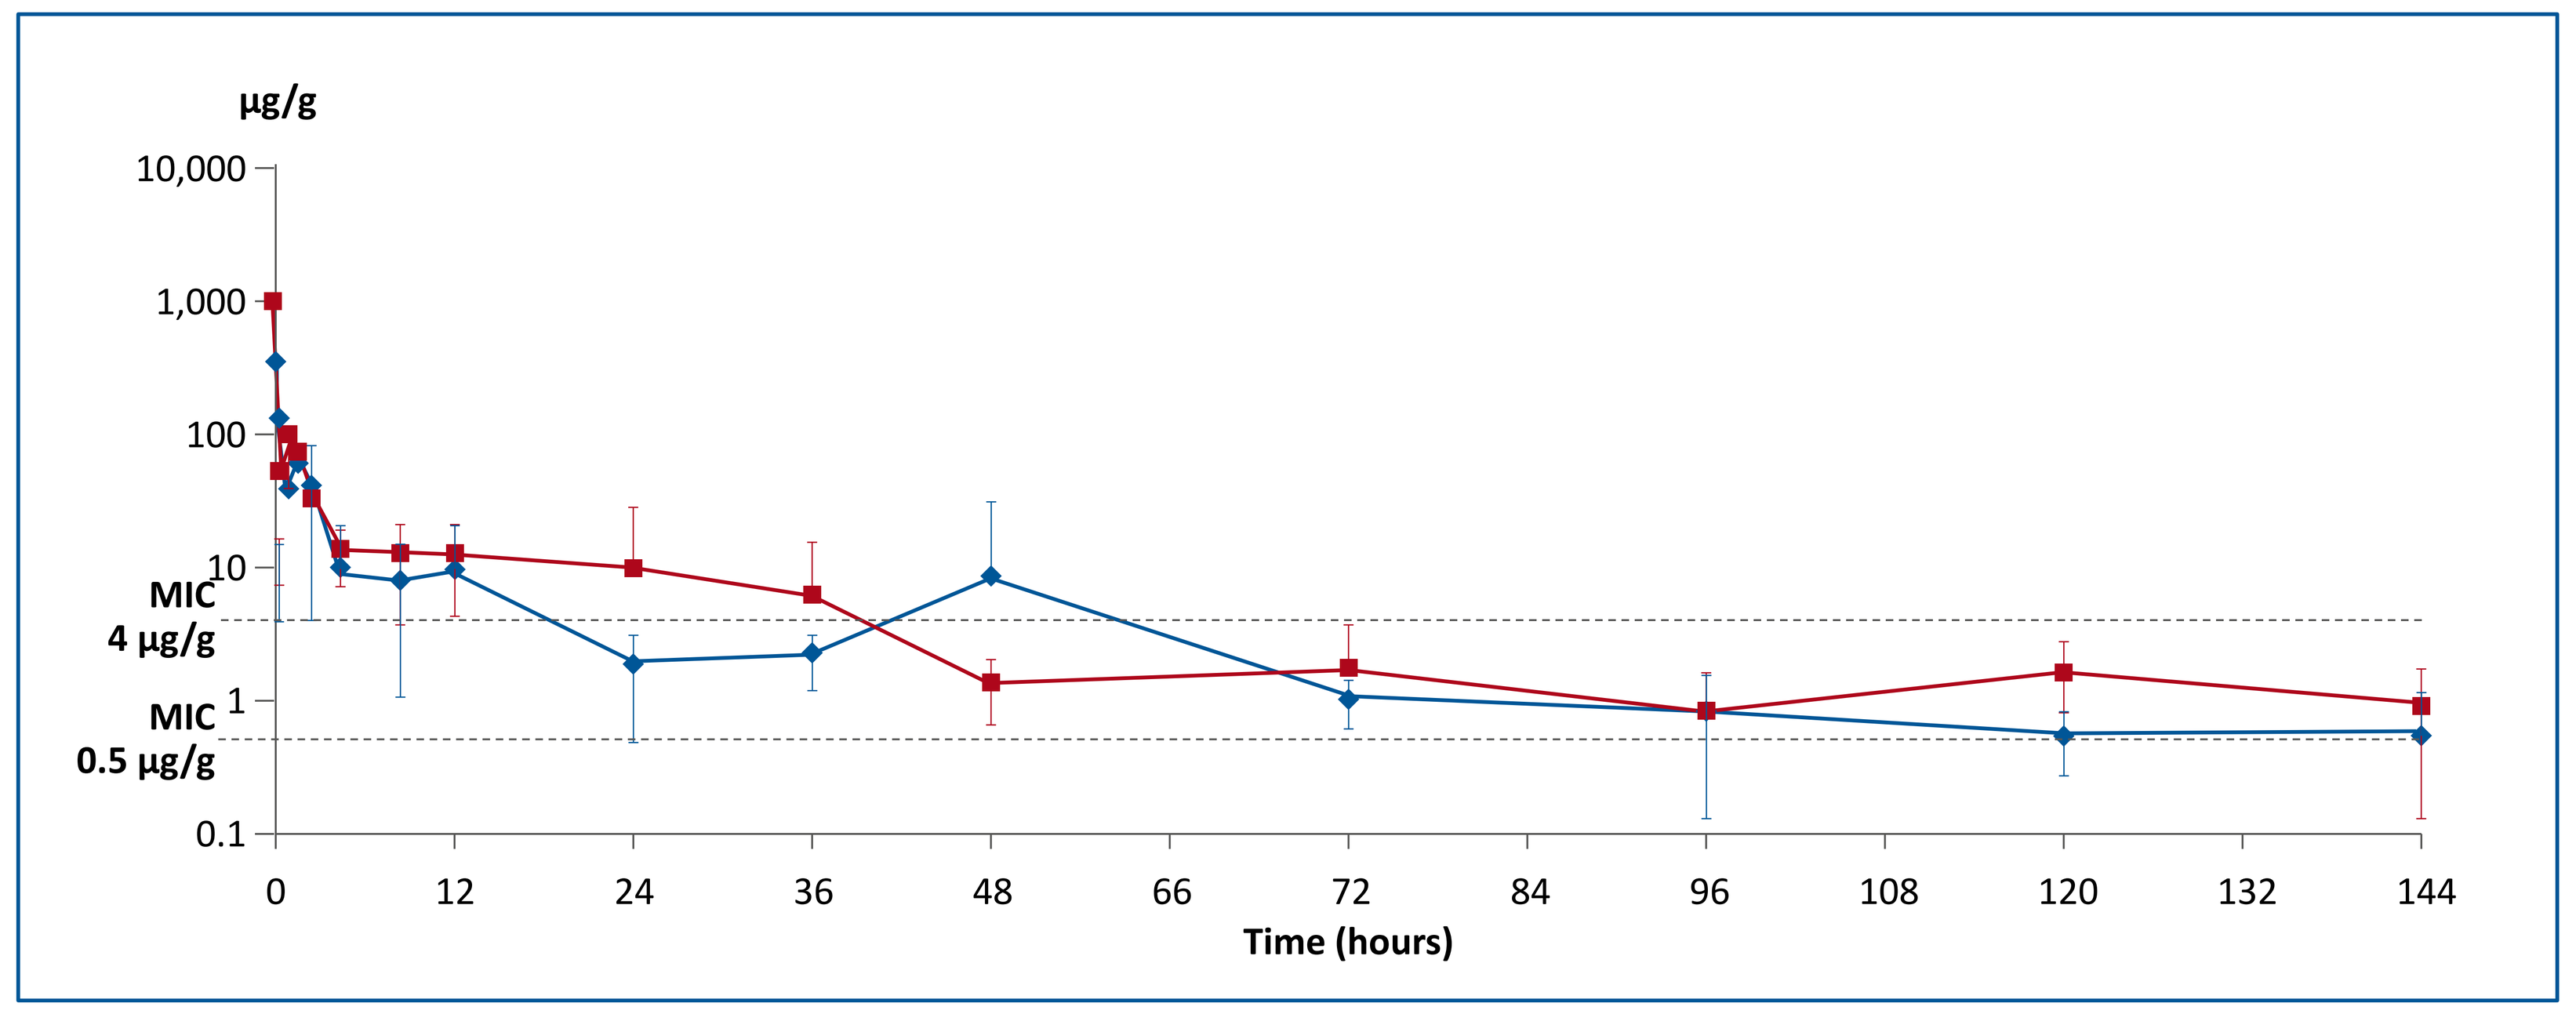
µg/g
10,000
1,000
100
10
MIC
4 µg/g
1
MIC
0.5 µg/g
0.1
0
12
24
36
48
66
72
84
96
108
120
132
144
Time (hours)

Figure 2: Azithromycin content in tears

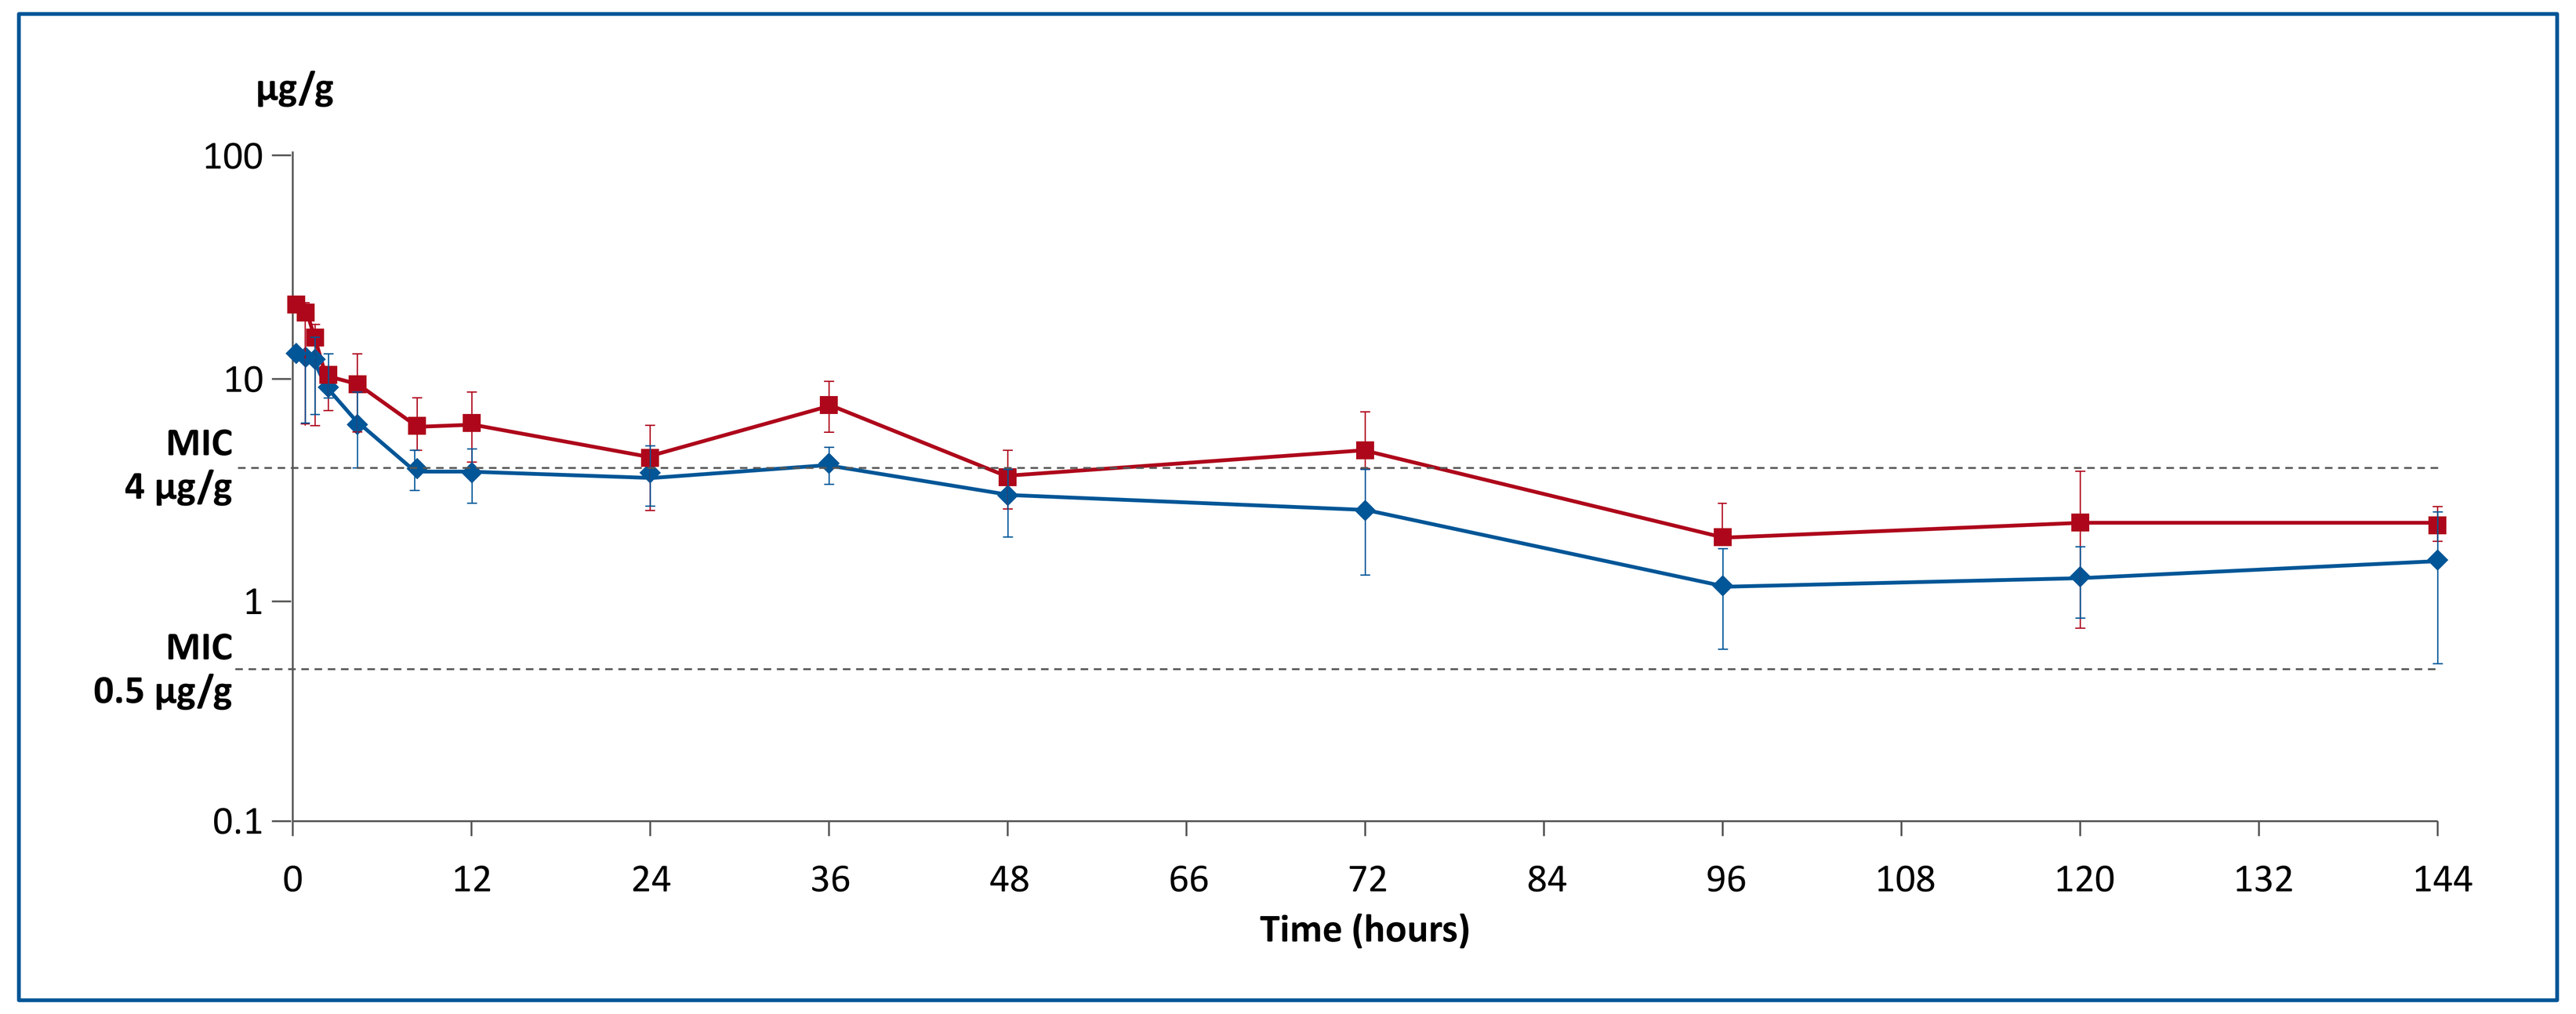
µg/g
100
10
MIC
4 µg/g
1
MIC
0.5 µg/g
0.1
0
12
24
36
48
66
72
84
96
108
120
132
144
Time (hours)

Figure 3: Azithromycin content in cornea

1.5% SFA-azithromycin 3% SFA-azithromycin

Results

Following a single topical instillation of 1.5% or 3% SFA-azithromycin in rabbit eyes, both formulations were macroscopically well tolerated. The highest exposure was found in the general order of eyelids > tears > cornea > bulbar conjunctiva > aqueous humor. The maximum concentrations of azithromycin were generally observed 5 minutes postdose, except for eyelids which showed a T_{max} of 1 hour. After an initial fast decline, in a second phase concentrations declined slowly over time with detectable concentrations at 144 hours postdose in all ocular tissues (Figures 1–3). Clinically relevant concentrations were maintained over 24 hours in eyelids, tears and cornea. Based on the AUC, azithromycin exposure in tissues was 1.84 to 2.70 times higher for the 3% SFA-azithromycin than the 1.5% azithromycin formulation.
The obtained values were compared with data from a similar experiment of a marketed 1.5% azithromycin formulation in medium-chain triglycerides[1], with a single instillation of 25 µL. The comparison showed that azithromycin exposure after administration of 1.5% SFA-azithromycin formulation was similar to the marketed formulation despite the lower absolute dose. In addition Amar et al[1] used, based on a literature search of the most common causative gram-positive and negative bacteria, a minimum inhibitory concentration (MIC) range of 0.5–4 µg/g for interpreting the ocular pharmacokinetic results. The SFA-azithromycin levels up to 144 hours postdose in eyelids, tears and cornea were above the MIC of 0.5 µg/g and for eyelids even above 4 µg/g.

The preservative-free, multi-dose SFA-azithromycin formulation was well tolerated. Both concentrations resulted in adequate and long-lasting azithromycin levels in tear film and eyelids, cornea and also conjunctiva. Based on these results the SFA based azithromycin formulations may lead to a new well-tolerated therapeutic for treating ocular surface bacterial infections such as blepharitis, conjunctivitis, and keratitis.

Reference
1: Amar et al, Current Eye Research, 33: 149-158, 2008

Financial disclosure of the authors:
K. Fischer, R. Grillenberger, S. Krösser (Novaliq employees); T. Amar (IRIS Pharma employee)

iris PHARMA